Bioenergetic profiling of cancer cell lines: quantifying the impact of glycolysis on cell proliferation.

Natalia Romero, Pamela M. Swain, Yoonseok Kam, George Rogers, Brian P. Dranka

Cell Analysis Division, Agilent Technologies, Lexington, MA

AACR 2018
POSTER # 3487

Agilent
Trusted Answers

Introduction

- Fast proliferating cells require tight regulation between the use of nutrients for ATP production (through glycolysis and oxidative phosphorylation) and the use of intermediate metabolites to sustain the increased biosynthetic activity.
- Cancer cells exhibit high glycolytic activity during rapid proliferation even in the presence of normal oxygen concentrations in culture. However, the role of glycolysis is not necessary as a major contributor of ATP but to allow nutrient assimilation into biosynthetic precursors.
- In this study we evaluated the contribution of the two main cellular metabolic pathway to ATP production rate in a panel of representative 18 cancer cell lines (10 of them from NCI-60 panel) and 2 control cell lines.

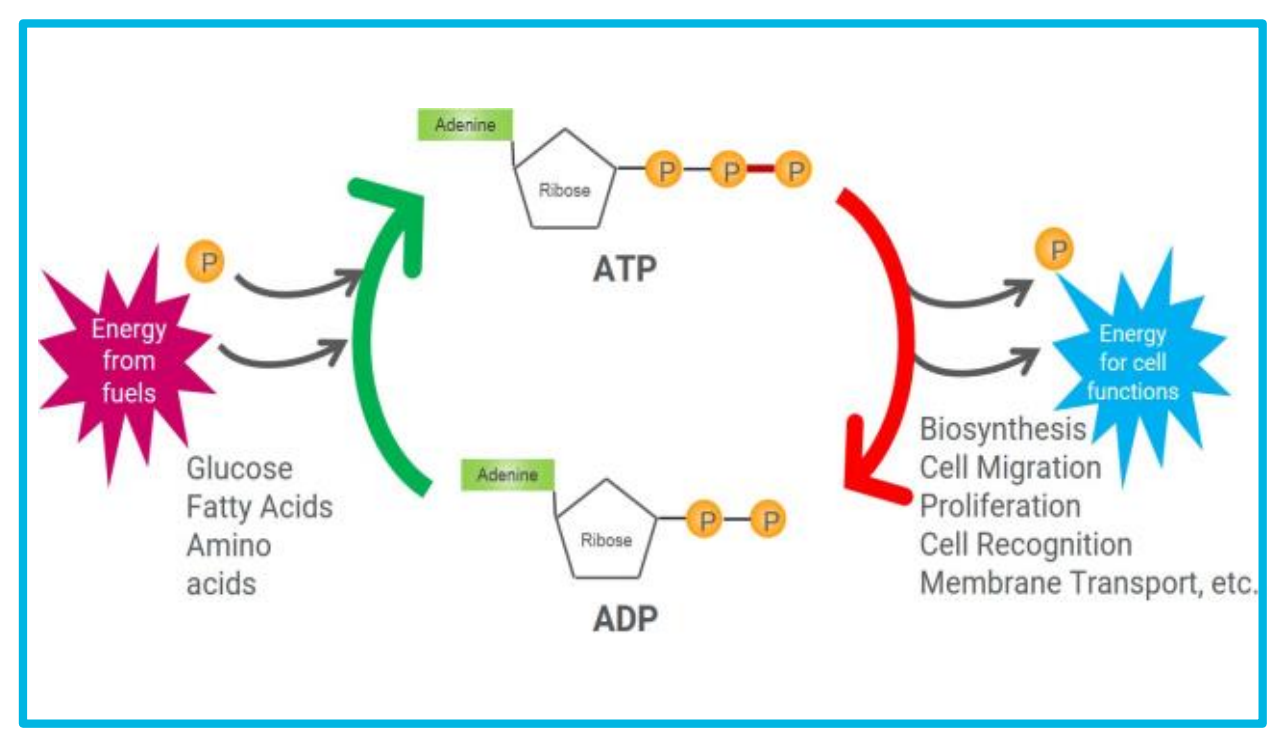
Adenine
Ribose
ATP
P
Energy from fuels
Glucose
Fatty Acids
Amino acids
ADP
Energy for cell functions
Biosynthesis
Cell Migration
Proliferation
Cell Recognition
Membrane Transport, etc.

Figure 1: ATP is the universal source of energy for every cellular function. ATP production and demand are tightly regulated to maintain intracellular ATP levels constant.

Experimental

Method

The Agilent Seahorse XF Real-Time ATP Rate Assay is a cell-based assay which allows simultaneous measurement of the two-main bioenergetic pathways to calculate the total rate of cellular ATP production as well as the fractional contribution from each pathway.

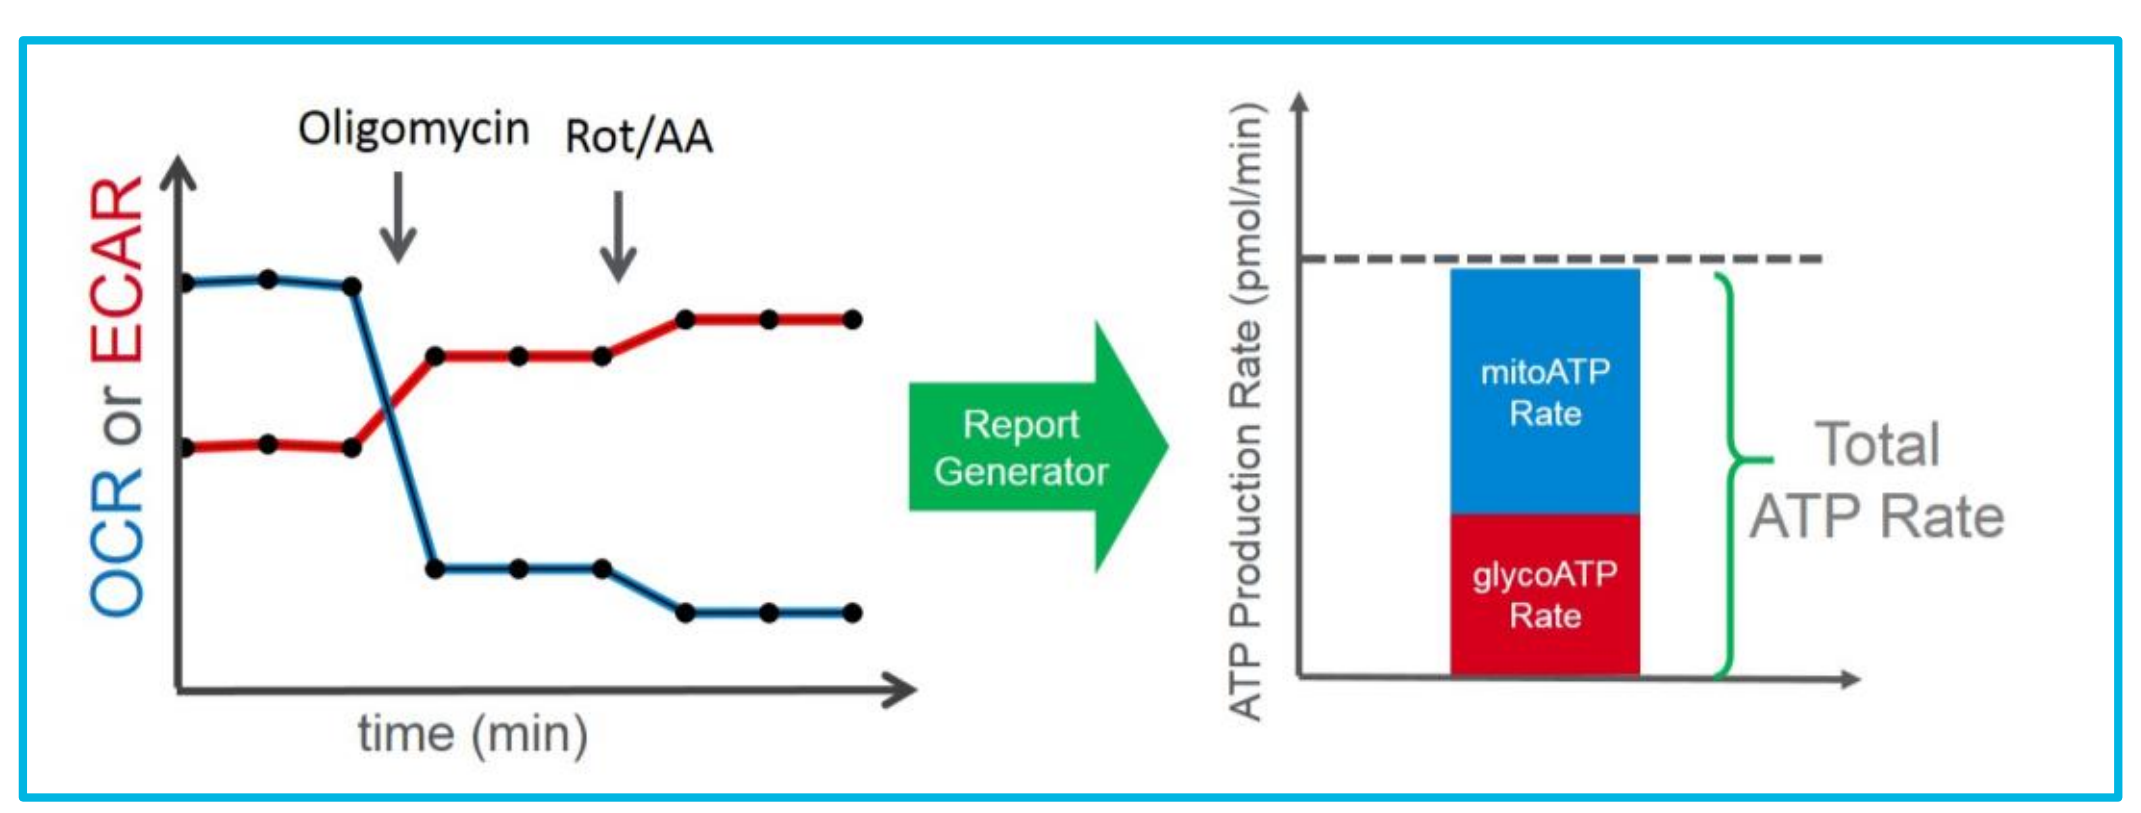
Oligomycin
Rot/AA
OCR or ECAR
time (min)
Report Generator
ATP Production Rate (pmol/min)
mitoATP Rate
glycoATP Rate
Total ATP Rate

Figure 2. Representative scheme of Agilent Seahorse XF Real-Time ATP Rate Assay. Kinetic profile of oxygen consumption rate (OCR) and extracellular acidification rate (ECAR) measurement. OCR and ECAR rates are first measured. Injection of oligomycin results in an inhibition of mitochondrial ATP synthesis that results in a decrease in OCR, allowing mitochondrial ATP production rates to be quantified. Complete inhibition of mitochondrial respiration with rotenone plus antimycin A enables calculation of mitochondrial-associated acidification, allowing calculation of glycolytic ATP production.

Results and Discussion

Use of XF Real-Time ATP Rate assay to study fuel dependencies for ATP Production

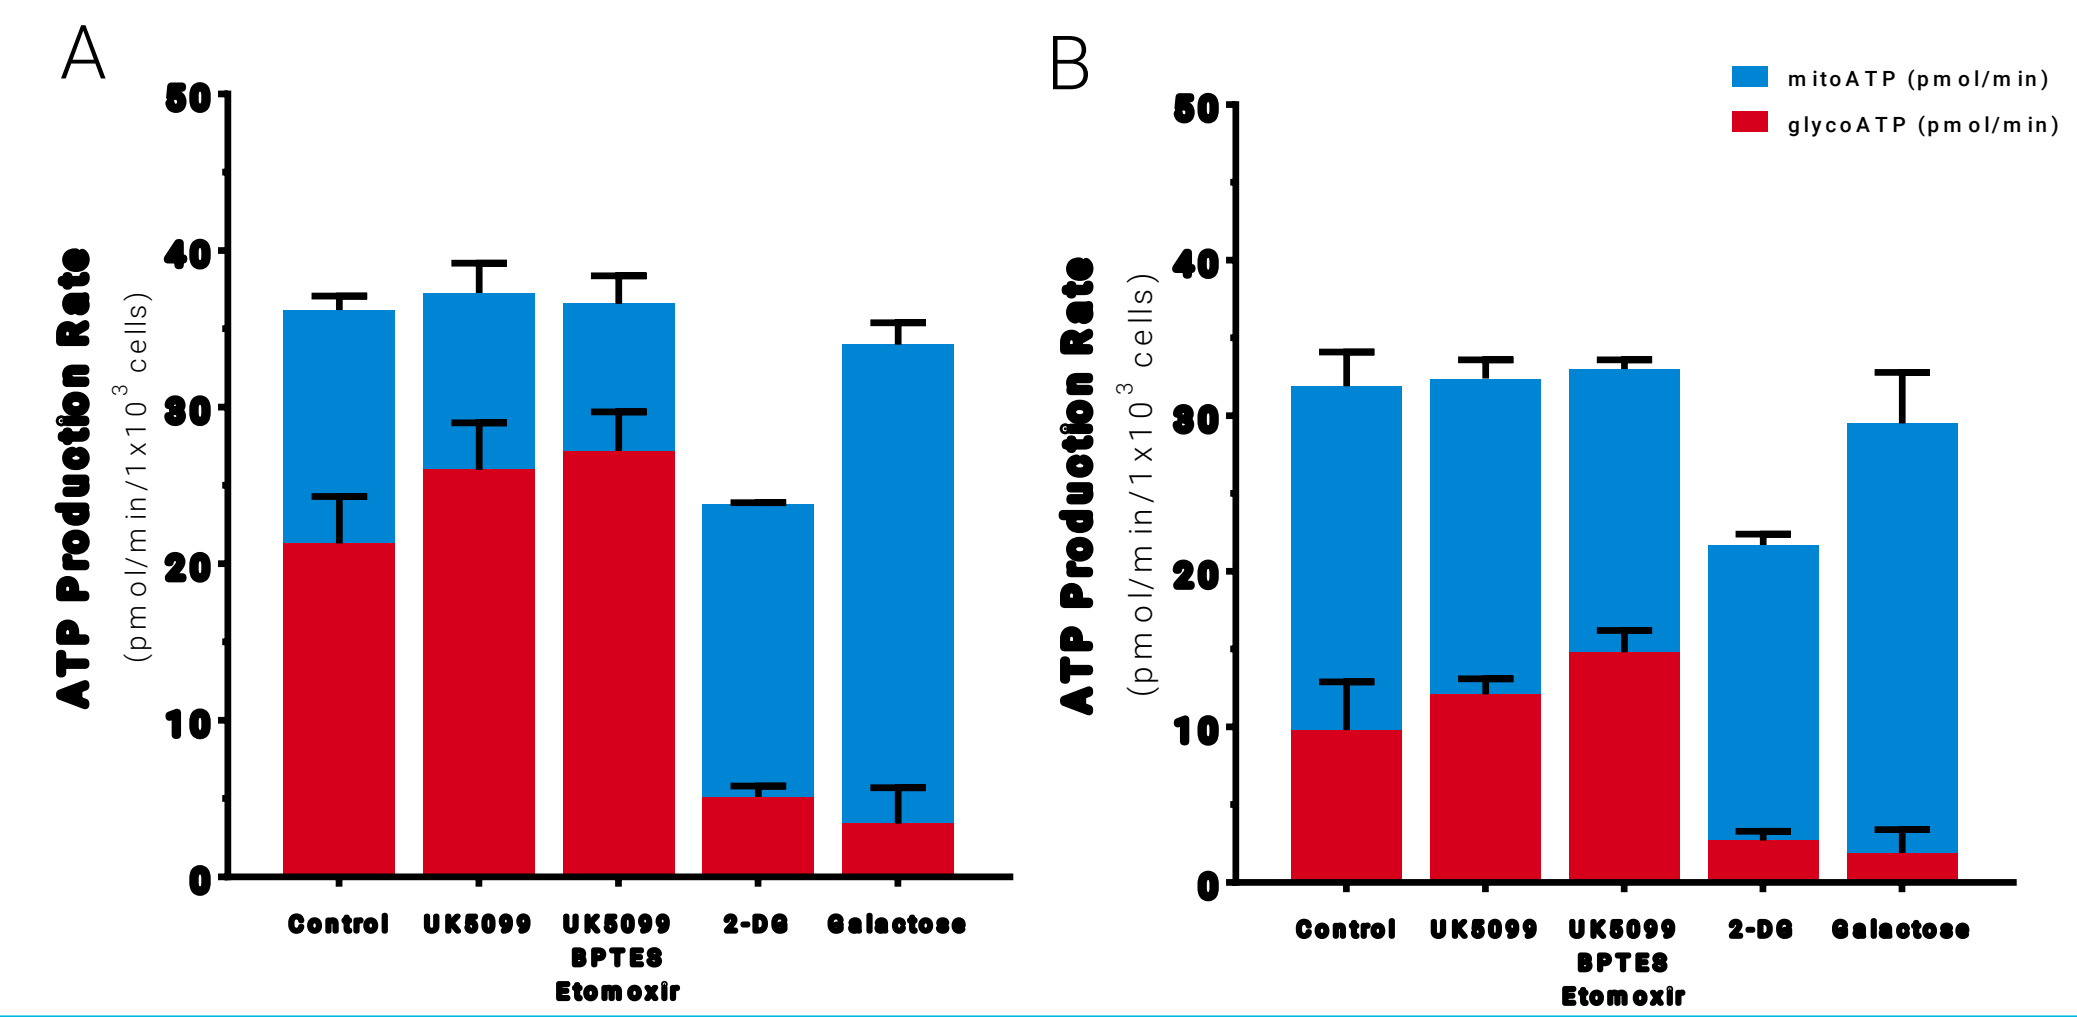
A
B
mitoATP (pmol/min)
glycoATP (pmol/min)
ATP Production Rate
(pmol/min/1x10³ cells)
Control
UK5099
UK5099 BPTES Etomoxir
2-DG
Galactose

Figure 3: XF Real-Time ATP Rate Assay was performed in A431 cells (A) and MCF7 cells (B) in the presence of the metabolic inhibitors UK5099 (inhibitor of pyruvate metabolism), BPTES (glutaminase inhibitor), Etomoxir (long chain fatty acid oxidation inhibitor), 2-DG (glycolytic inhibitor) or replacing glucose by galactose in the extracellular medium.

Contribution of glycolysis to ATP production is very variable between representative cancer cell lines

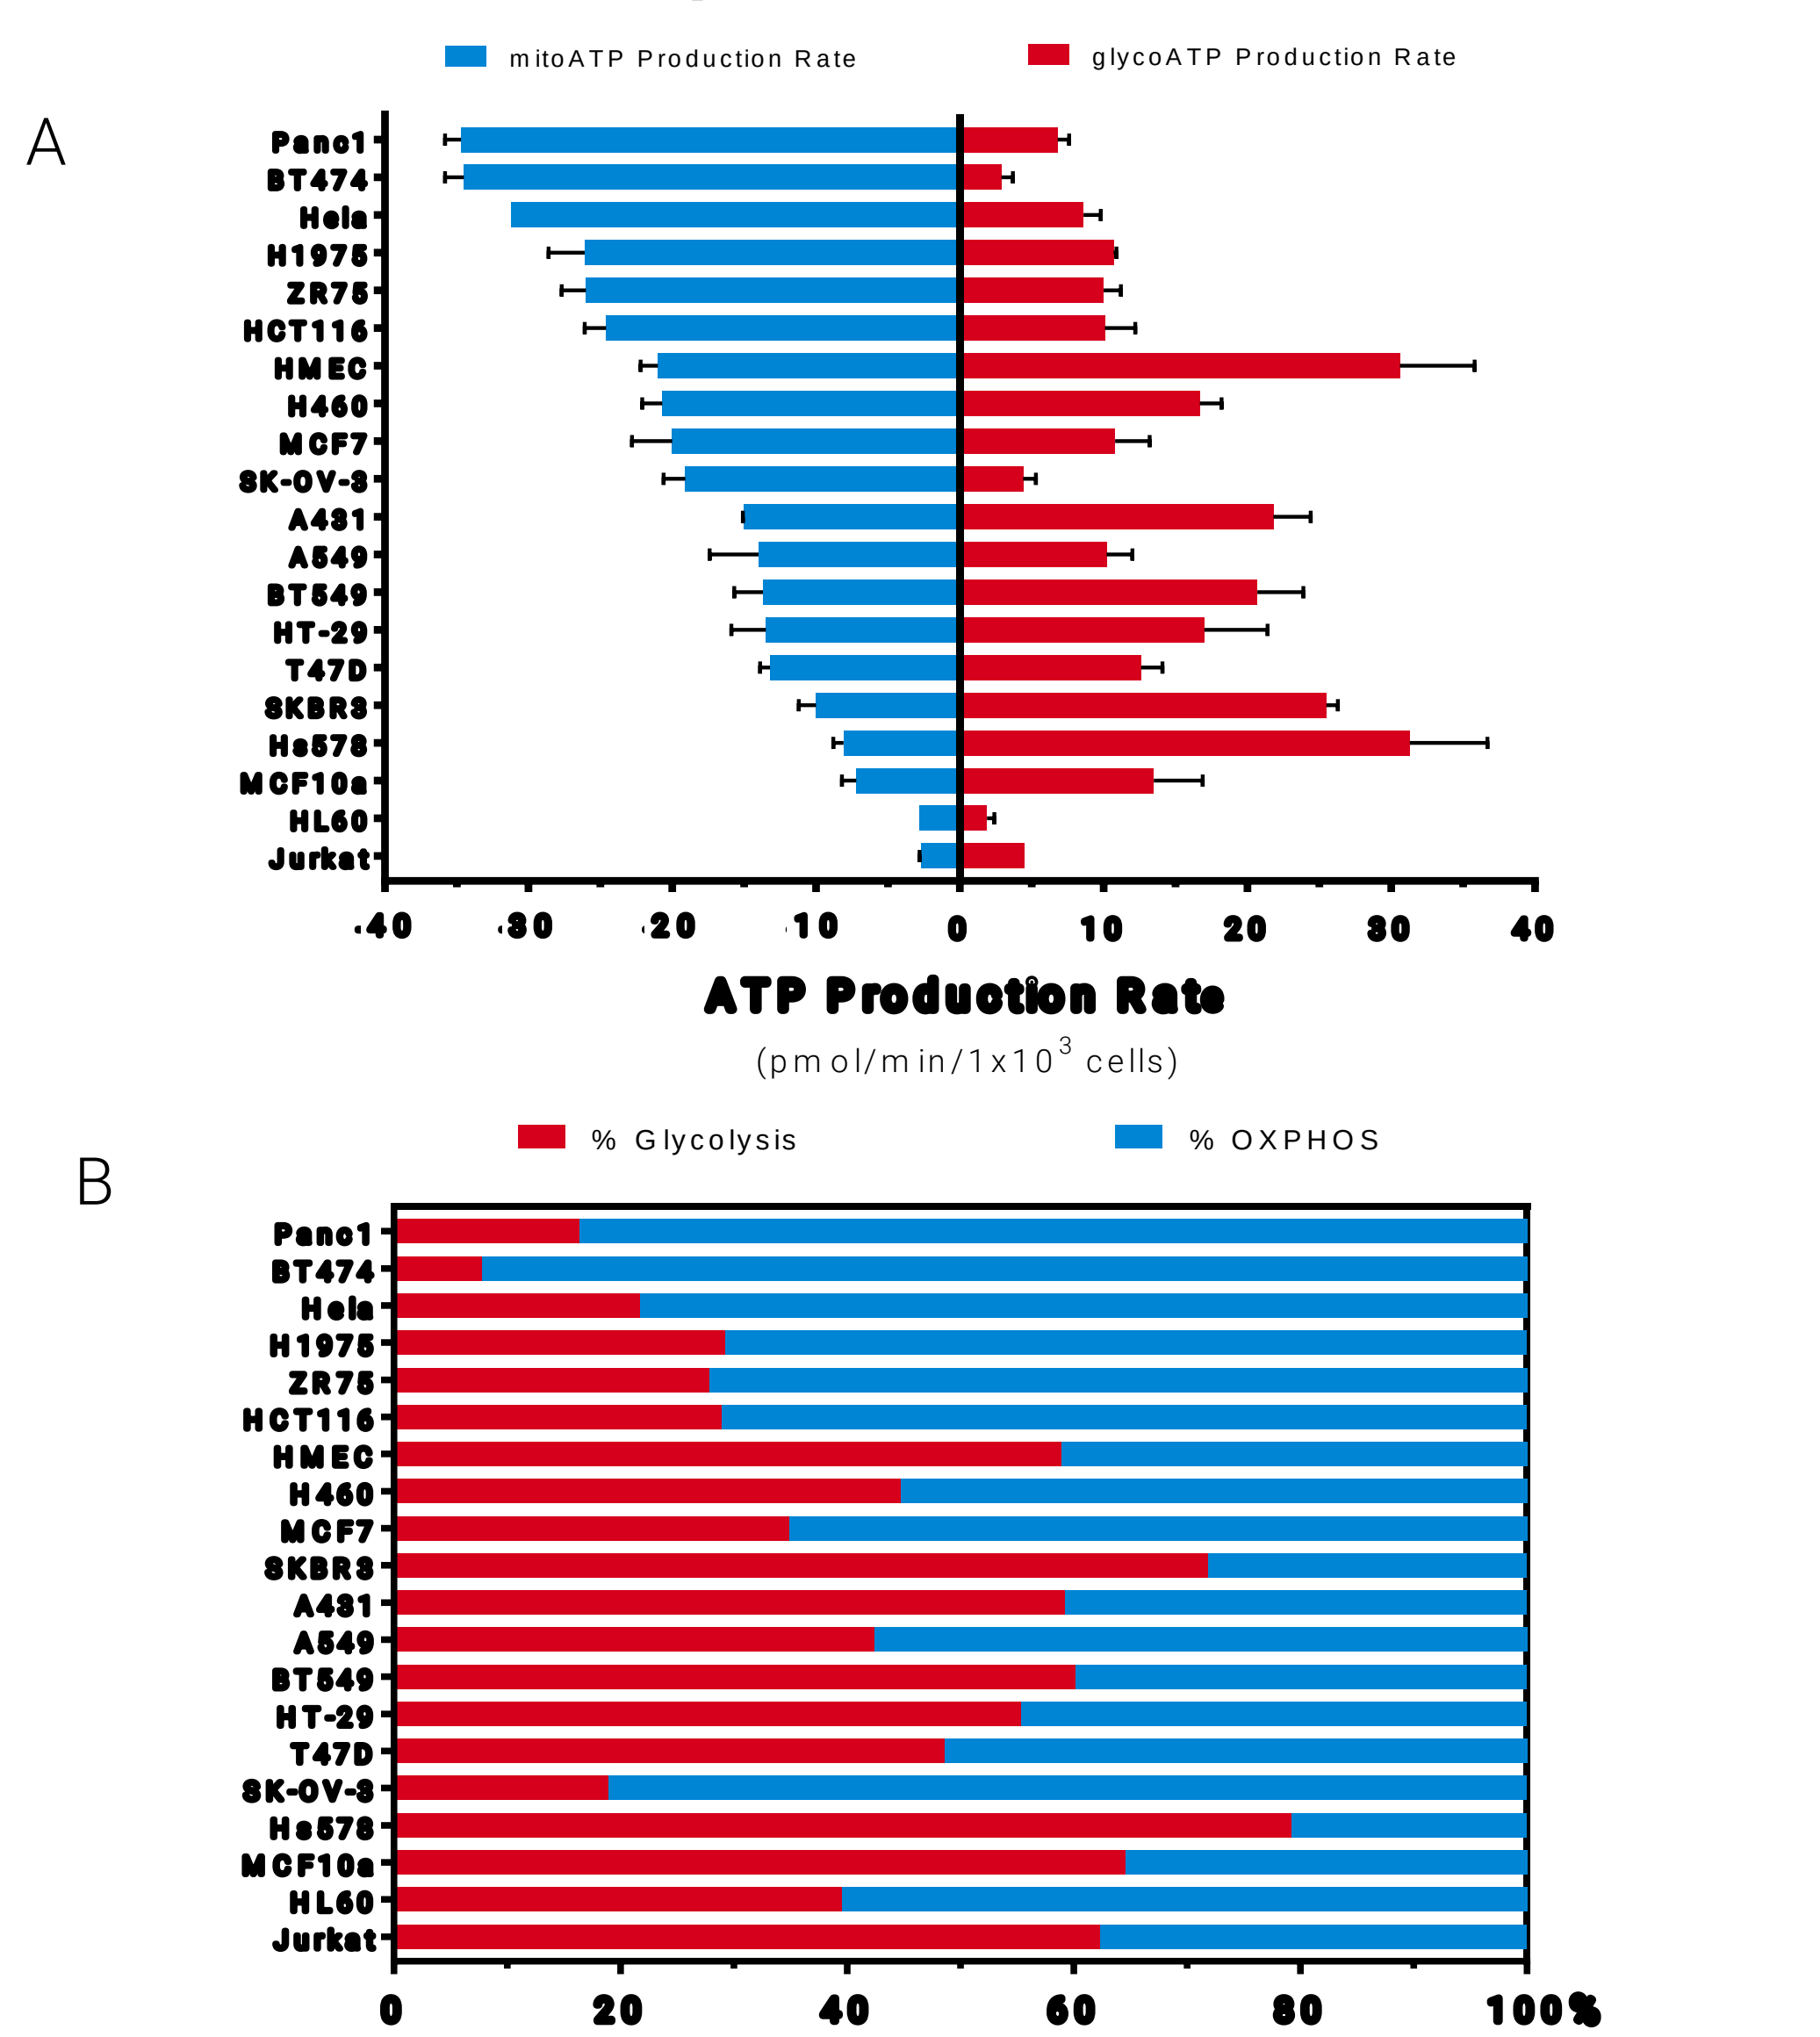
mitoATP Production Rate
glycoATP Production Rate
A
Panc1
BT474
Hela
H1975
ZR75
HCT116
HMEC
H460
MCF7
SK-OV-3
A431
A549
BT549
HT-29
T47D
SKBR3
Hs578
MCF10a
HL60
Jurkat
ATP Production Rate
(pmol/min/1x10³ cells)
B
% Glycolysis
% OXPHOS
100%

Figure 4: (A) Glycolytic and mitochondrial ATP production rate distribution in a representative panel of cancer cells lines. (B) Relative contribution of ATP production from glycolysis and oxidative phosphorylation to total ATP Production.

Results and Discussion

Mitochondrial and total ATP production rates inversely correlate with cell proliferation rate, but not glycolytic ATP production rate

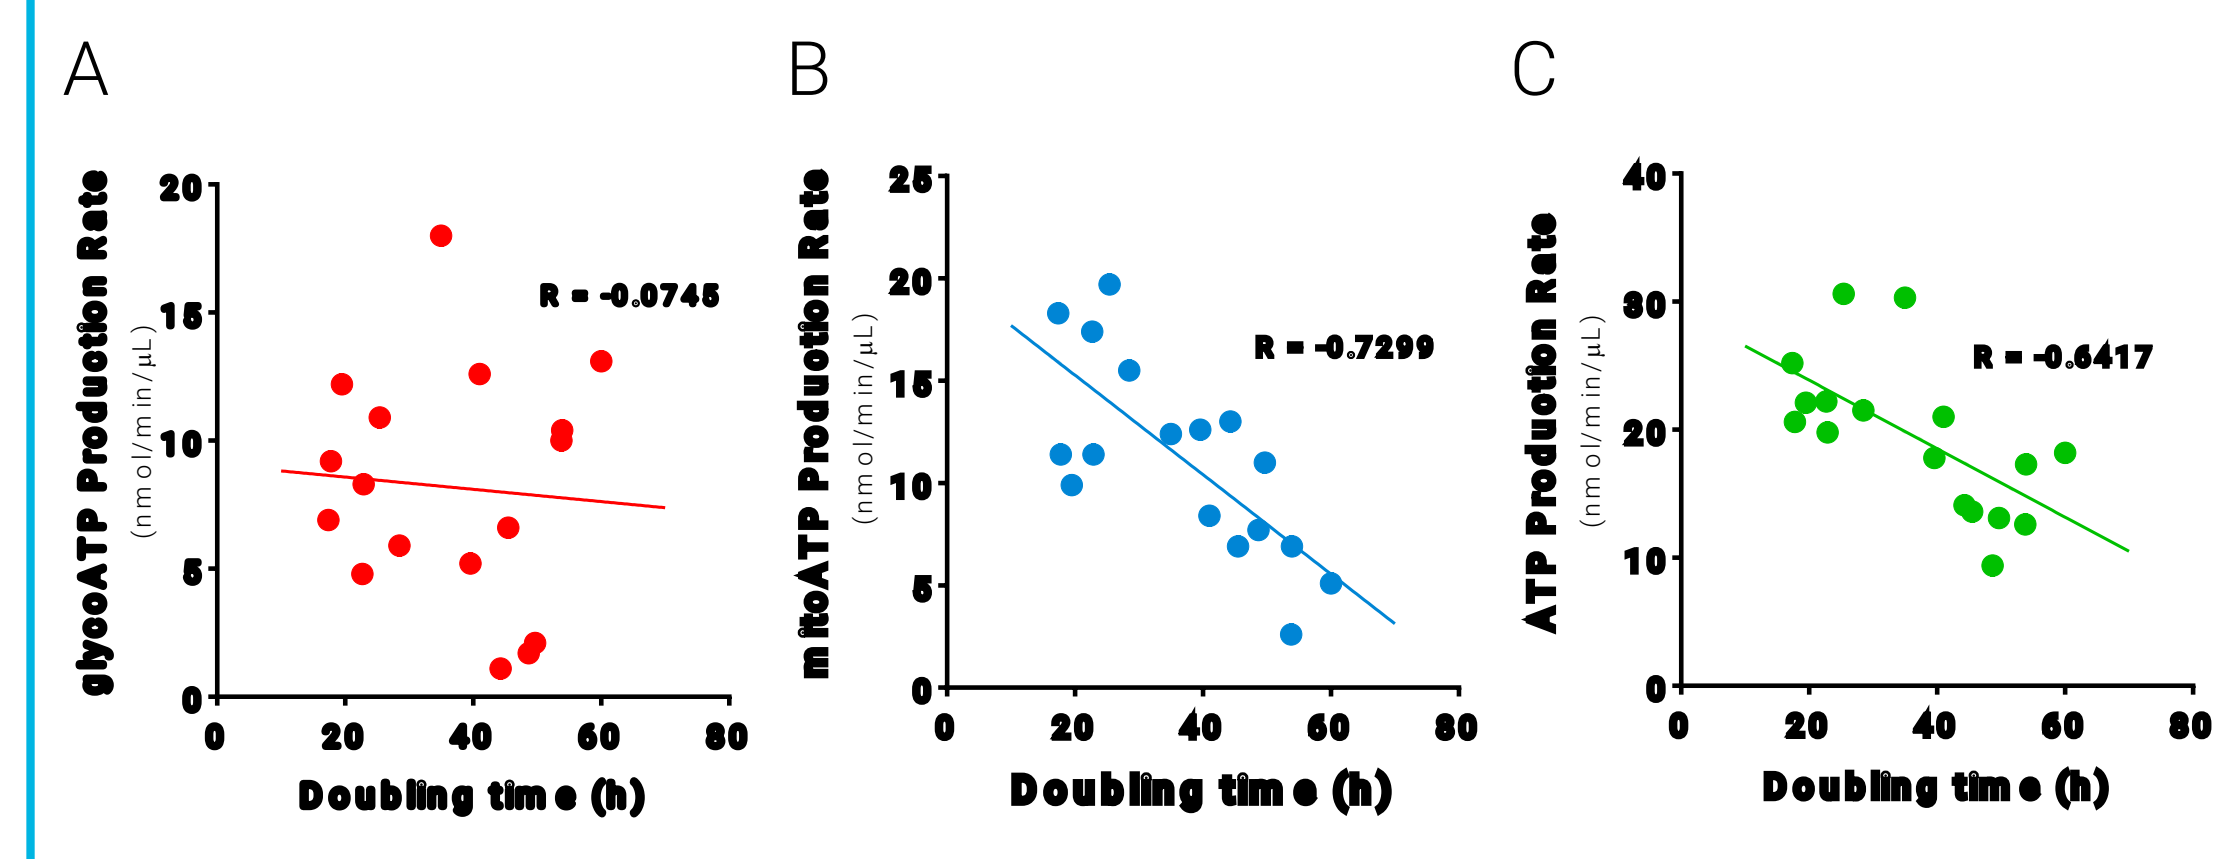
A
B
C
glycoATP Production Rate
mitoATP Production Rate
ATP Production Rate
(nmol/min/µL)
R = -0.0745
R = -0.7299
R = -0.6417
Doubling time (h)

Figure 5: (A) glycoATP production rate (B) mitoATP production rate and (C) total ATP production rate were calculated for the panel of cancer and control cells of Fig. 4 using Seahorse XF Real-Time ATP Rate Assay. Rates are expressed as pmol/min/µL of cellular cell volume.

Breast cancer cell lines ER+ have a more oxidative metabolism than ER- or control cell lines

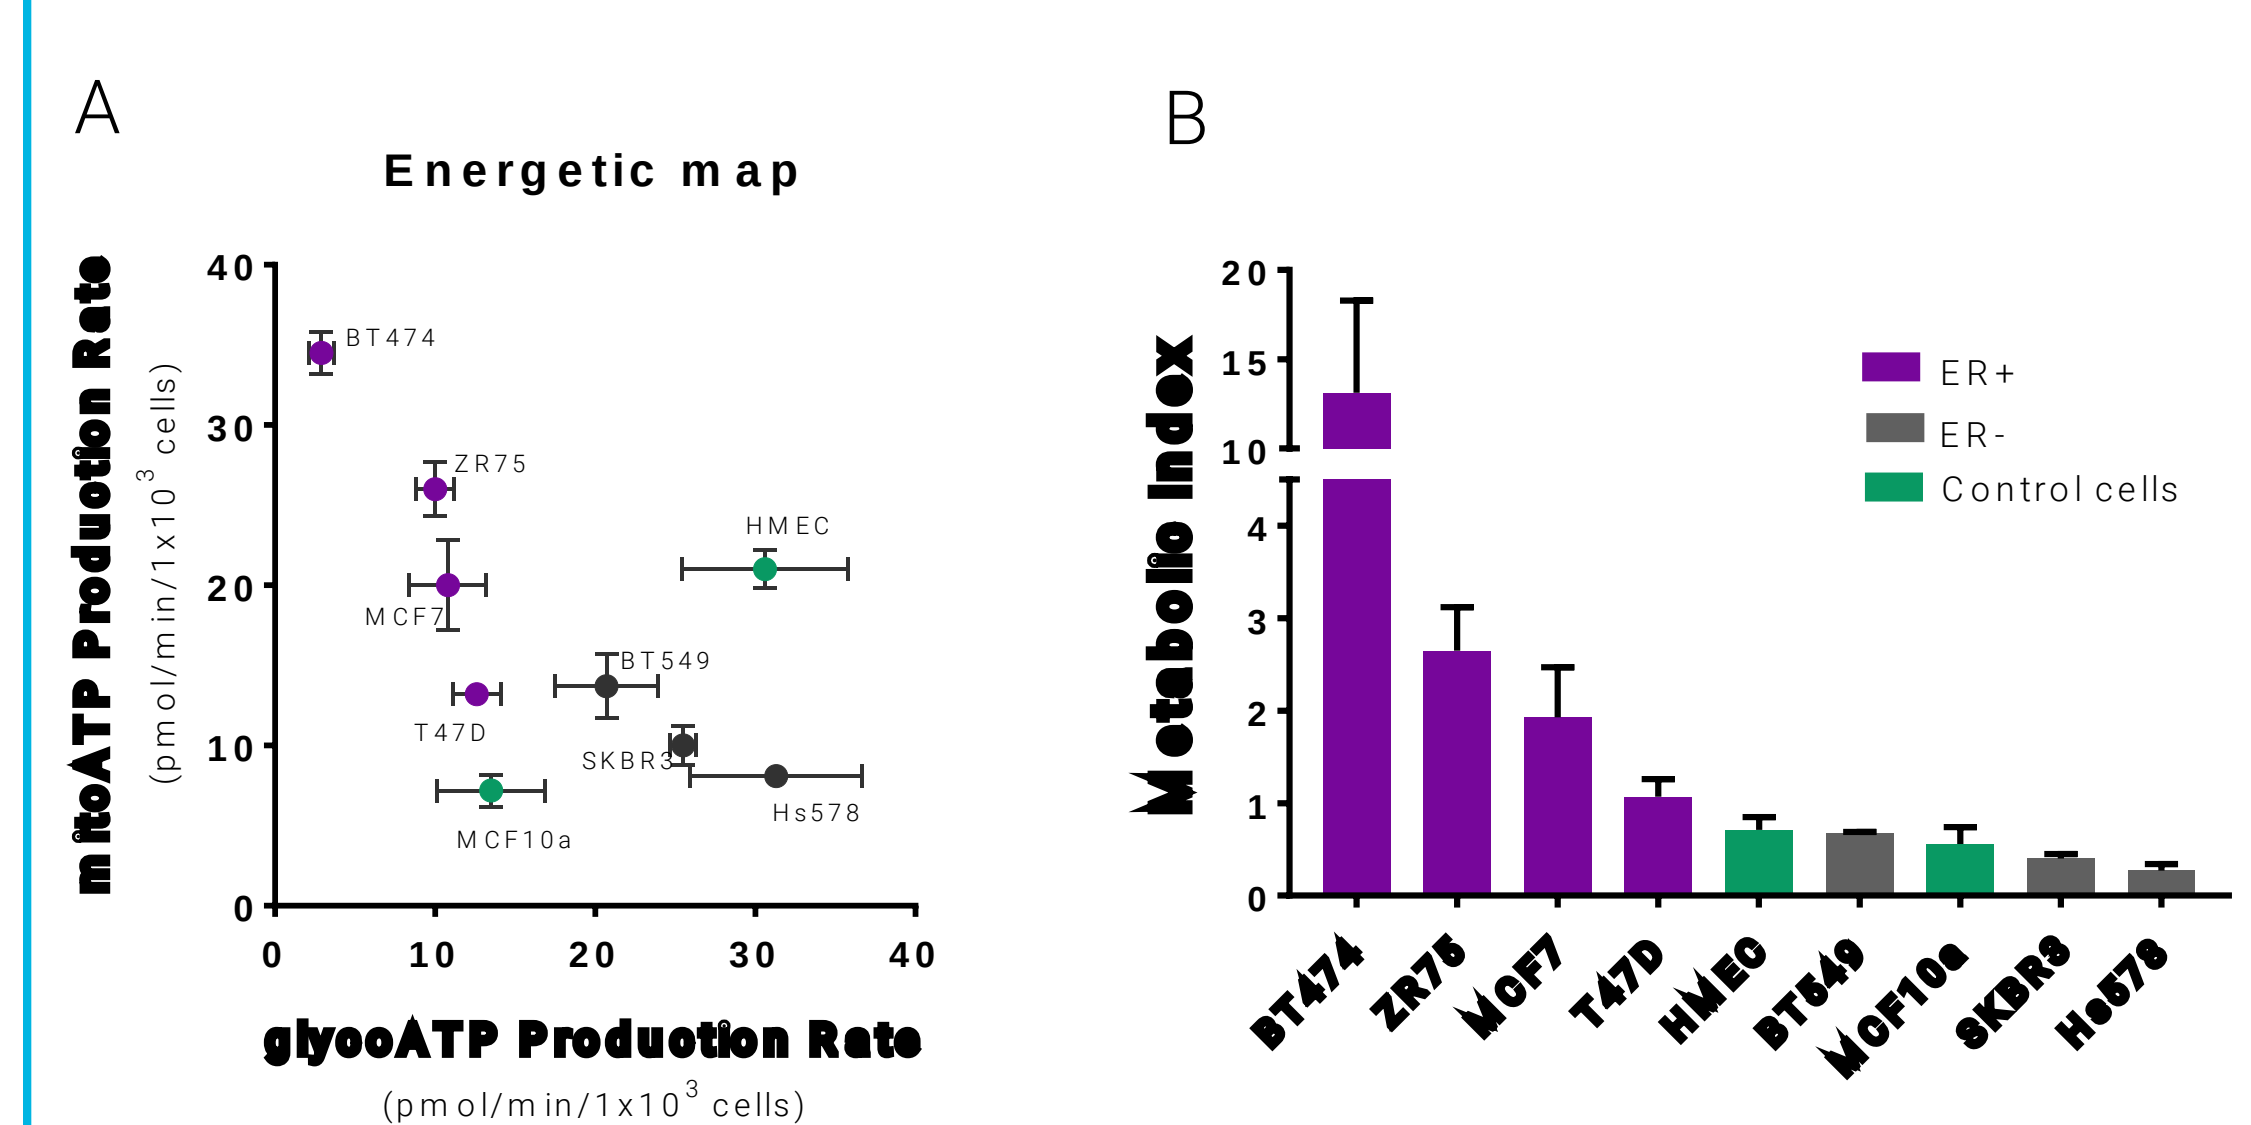
A
Energetic map
mitoATP Production Rate
(pmol/min/1x10³ cells)
BT474
ZR75
MCF7
HMEC
BT549
T47D
SKBR3
Hs578
MCF10a
glycoATP Production Rate
(pmol/min/1x10³ cells)
B
Metabolic Index
ER+
ER-
Control cells

Figure 6: Evaluation ATP Production Rate in a breast cancer cell panel. (A) Energy map showing the distribution of mitoATP production rate vs glycoATP production rate across the 7 cancer cell lines and 2 normal breast-derived cell lines. (B) Metabolic index (mitoATP Production Rate/glycoATP Production Rate) of the breast cancer lines analyzed showing that estrogen receptor positive (ER+) cancer cell lines analyzed in this study have higher metabolic index.

Conclusions

- The new Seahorse XF Real-Time ATP Rate Assay allows the measurement of total ATP production rate in cells as well as to distinguish between the fraction coming from mitochondrial oxidative phosphorylation or from glycolysis, the two main metabolic pathways responsible for ATP production in mammalian cells.
- The XF Real-Time ATP Rate Assay can be used to study cellular dependencies of metabolic fuels for ATP production as well as to characterize compounds that induce metabolic switches without affecting total energy production.
- Using a representative panel of frequently used cancer cell lines we found that even in cells typically considered highly glycolytic, the ATP production from glycolysis is only between 30-60% of total ATP production.
- mitoATP and total ATP production rates, but not glycoATP production rate inversely correlate with cell doubling times in the 17 adherent cells of the cancer panel analyzed. This result highlights the role of mitochondrial OXPHOS as energy supplier for cell proliferation.
- The panel of breast cancer cells analyzed shows wide variations in the metabolic phenotype of the individual cell lines. Interestingly, subgroup of ER+ breast cancer lines showed a higher metabolic index indicative of higher reliance on mitoATP production in these cells. The significance of this correlation will require further studies.
- The developed assay represents a completely new tool for improved characterization of the bioenergetic profile of cancer cell variants and will allow to better understand the role of energy metabolism in cancer cell biology.

References

Zu, X.L., and Guppy, M. Cancer Metabolism: facts fantasy and fiction. *Biochem. Biophys. Res. Comm*. 313:459-465. (2004)

Ward, P.S. and Thomson, C.B. Metabolic Reprogramming: A cancer hallmark even Warburg did not anticipate. Cancer Cell 21: 297-308 (2012).

https://www.agilent.com/en/promotions/seahorse-xf-technology

For Research Use Only. Not for use in diagnostic procedures